A phactMI™ Benchmarking Survey: 27 Medical Information Departments on Healthcare Decision Making Materials for Payer Inquiries

AnnaMaria D'Ascoli[1] RPh, PharmD, MBA; Andrew Gazo[2] RPh, PharmD; and Sue Yueh[3], RPh, Pharm D, MBA, on behalf of the phactMI™ Benchmarking Committee

[1] Director, Global Medical Information, Pfizer Inc., New York, NY [2] Head of Global Content Sanofi, Bridgewater, NJ & phactMI Board Member [3] Associate Director, Global Medical Information, Celgene Corporation, Summit, NJ

phactMI
Pharma Collaboration for Transparent Medical Information

BACKGROUND

Medical Information departments (MIDs) in pharmaceutical companies respond to unsolicited requests from health care decision makers (HCDMs) using various materials. These materials are created under the guidance of Academy of Managed Care Pharmacy (AMCP) which essentially governs how some of these materials are written and formatted. With the evolving healthcare landscape, the needs of HCDMs have also changed which MIDs should be prepared for.

OBJECTIVE

The objective of this survey was to evaluate how 27 MIDs respond to payer requests for information, and to identify payer needs in order to provide a better overall customer experience.

METHODS

- This 16-question survey administered between Dec 12, 2017 and Feb 28, 2018 collected information from 27 MIDs about their management of payer inquiries.
- Questions ranged from yes or no, to multiple choice (select all that apply), or open ended (open text field). Data was compiled, organized into bar graphs and pie charts, and analyzed for results.

RESULTS

The results from this survey provided insight into the management of payer unsolicited medical requests (PUMRs) and HCDM materials used by MIDs when responding to inquiries. The data collected compared 27 MIDs to evaluate organizational structure, resources, and management of materials employed in response to PUMRs.

Some Key Points from the Survey are as follows:

- Appropriate internal alignment with MI, health economics outcomes research (HEOR) and medical affairs (MA) are necessary for success with budgeting and planning for materials needed for HCDMs.
- Planning for HCDM materials 1-2 years prior to a product launch is key.
- Pre-approval material development in ~27% of companies using information primarily from published studies and/or summaries of submitted/planned pivotal studies.
- The majority of companies create AMCP dossiers for marketed products using an external agency and some MIDs post them using Dymaxium (e-dossier portal which allows only HCDMs to access). Not all MIDs provide payer materials online, partially due to expense, workload, and a low number of overall requests.

KEY FINDINGS FROM RESULTS

- Majority of PUMRs for dossiers are sent directly to MIDs from HCDMs or by field colleagues or commercial teams interacting with payers.

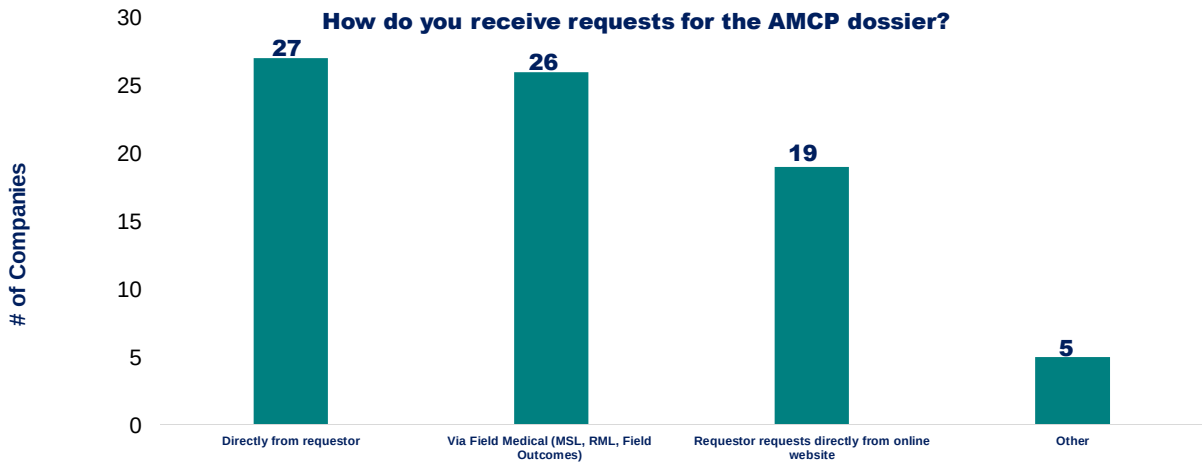
How do you receive requests for the AMCP dossier?
of Companies
30
25
20
15
10
5
0
27
26
19
5
Directly from requestor
Via Field Medical (MSL, RML, Field Outcomes)
Requestor requests directly from online website
Other

- The primary materials requested by payers from MIDs are the AMCP dossier (96%), followed by the budget impact model (81%), real world data (81%), and varying amounts of additional materials i.e. publications & posters (74%), comparative effectiveness data (62%), executive summaries (55%) & slide decks (51%).

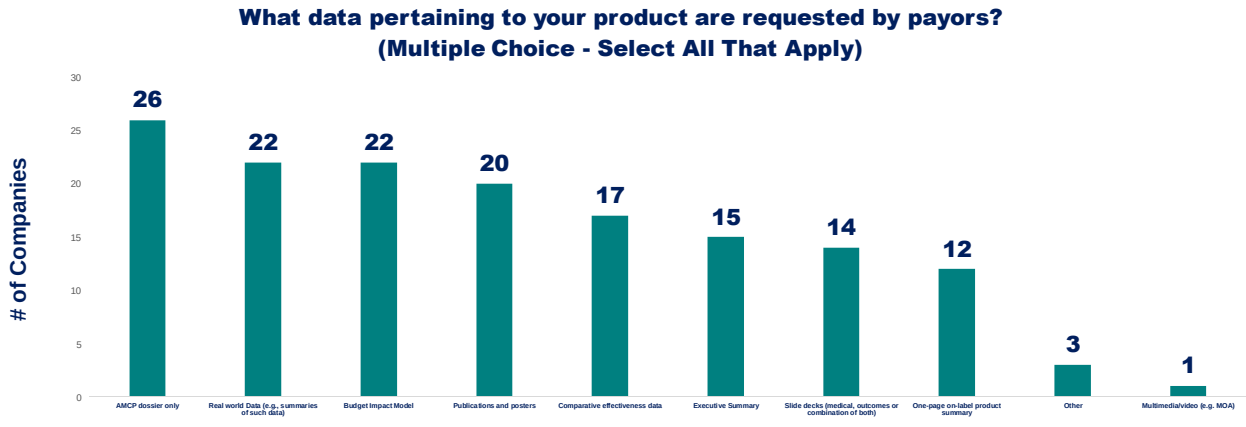
What data pertaining to your product are requested by payors?
(Multiple Choice - Select All That Apply)
of Companies
26
22
22
20
17
15
14
12
3
1
AMCP dossier only
Real world Data (e.g., summaries of such data)
Budget Impact Model
Publications and posters
Comparative effectiveness data
Executive Summary
Slide decks (medical, outcomes or combination of both)
One-page on-label product summary
Other
Multimedia/video (e.g. MOA)

- Most MIDs do not have dedicated staff to manage payer inquiries therefore each company should develop a method for resourcing MIDs depending on needs; some MIDs consider inquiry volume and expected launches, whereas others consider the product life cycle, payer management, or disease state complexity as factors.

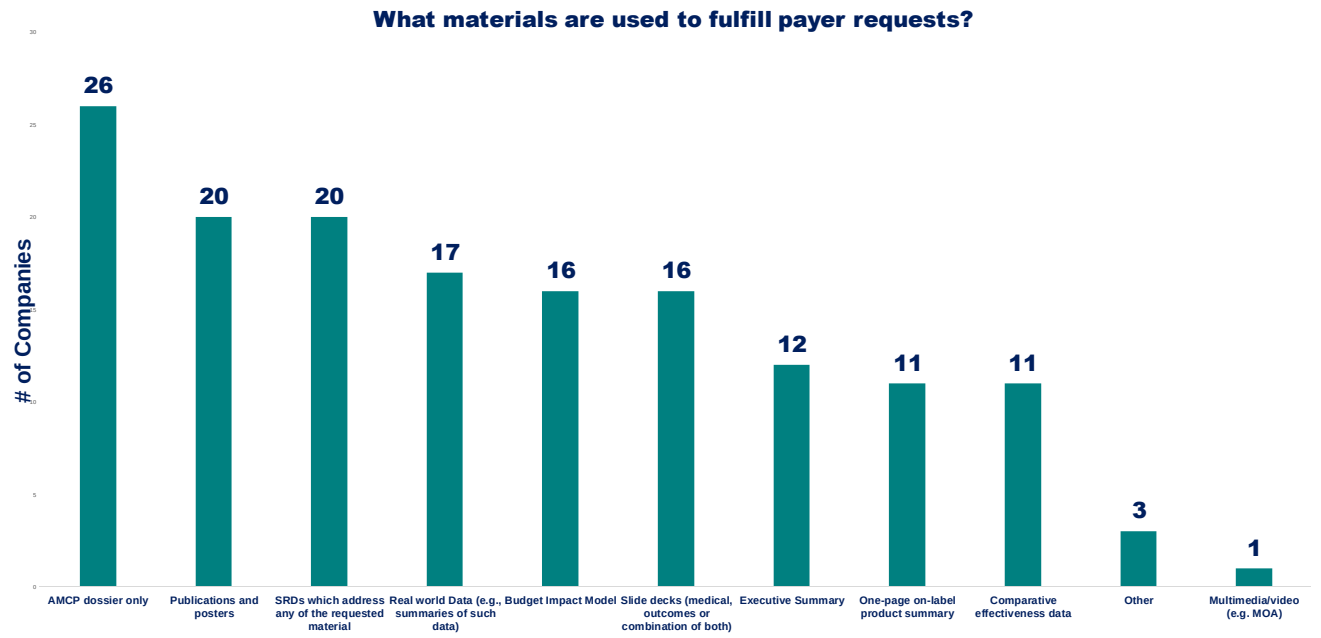
What materials are used to fulfill payer requests?
of Companies
26
20
20
17
16
16
12
11
11
3
1
AMCP dossier only
Publications and posters
SRDs which address any of the requested material
Real world Data (e.g., summaries of such data)
Budget Impact Model
Slide decks (medical, outcomes or combination of both)
Executive Summary
One-page on-label product summary
Comparative effectiveness data
Other
Multimedia/video (e.g. MOA)

- Majority of payer requests are fulfilled with AMCP dossiers (96%) (including e-dossier portals to help with easier online access & distribution) followed by publications and posters (74%) and customized standard response documents (76%).

DISCUSSION

Although there is no requirement for standardizing the management of PUMRs amongst MIDs, these results provide an opportunity for further discussion of several points on this topic, including the development of criteria for the posting of certain e-dossiers online versus others and reassessing the benefits of allowing online availability to certain health care decision making tools/materials versus others.

This survey highlighted areas of opportunities for MIDs to better prepare tools to address pipeline inquiries received from payers via field medical and commercial teams. Furthermore, the availability of preapproval materials is an area of much interest. We recommend further discussion to a systematic/consistent approach in creating tools to address types of PUMRs. Ultimately, it is the responsibility of the MID to develop appropriate, useful and accessible HCDM materials to efficiently respond to PUMRs. MIDs should be equipped with the resources and be prepared to deliver relevant information in a timely manner to meet the evolving needs of HCMDs.

Disclosures: The authors of this presentation have nothing additional to disclose concerning personal or financial relationships with commercial entities that may have direct or indirect interest in the subject matter of this presentation